P3914

E-LEARNING AND MENTORING TO FIGHT AGAINST ANTIMICROBIAL RESISTANCE

S.DAHO[1], M. POUPARD[1], M. KHALIFE[2], K.PYAR MIN HTIKE[3], J. KHRAIBANI[2], S.L.RUBAYAH[3], D. ABI HANNA[2], D. KIMATHI[3]

[1] MSF Academy for Healthcare, Médecins Sans Frontières, Brussels, Belgium
[2] MSF Academy for Healthcare, Médecins Sans Frontières, Beirut, Lebanon
[3] MSF Academy for Healthcare, Médecins Sans Frontières – Eastern Africa, Nairobi, Kenya

MEDECINS SANS FRONTIERES

MSF ACADEMY FOR HEALTHCARE

CONTACT DETAILS

BACKGROUND

MSF Academy for Healthcare and the British Society for Antimicrobial Chemotherapy (BSAC) developed the Antimicrobial Resistance (AMR) Learning Intiatitive with 2 online diploma courses:

- **Antimicrobial Stewardship** (AMS),
- **Infection Prevention and Control** (IPC) Supervision and Management

This initiative was created to respond to the AMR global challenges, by strengthening capacities of the AMS & IPC professionals working in MSF-supported healthcare facilities located mostly in low and middle incomes countries, including in conflict areas.

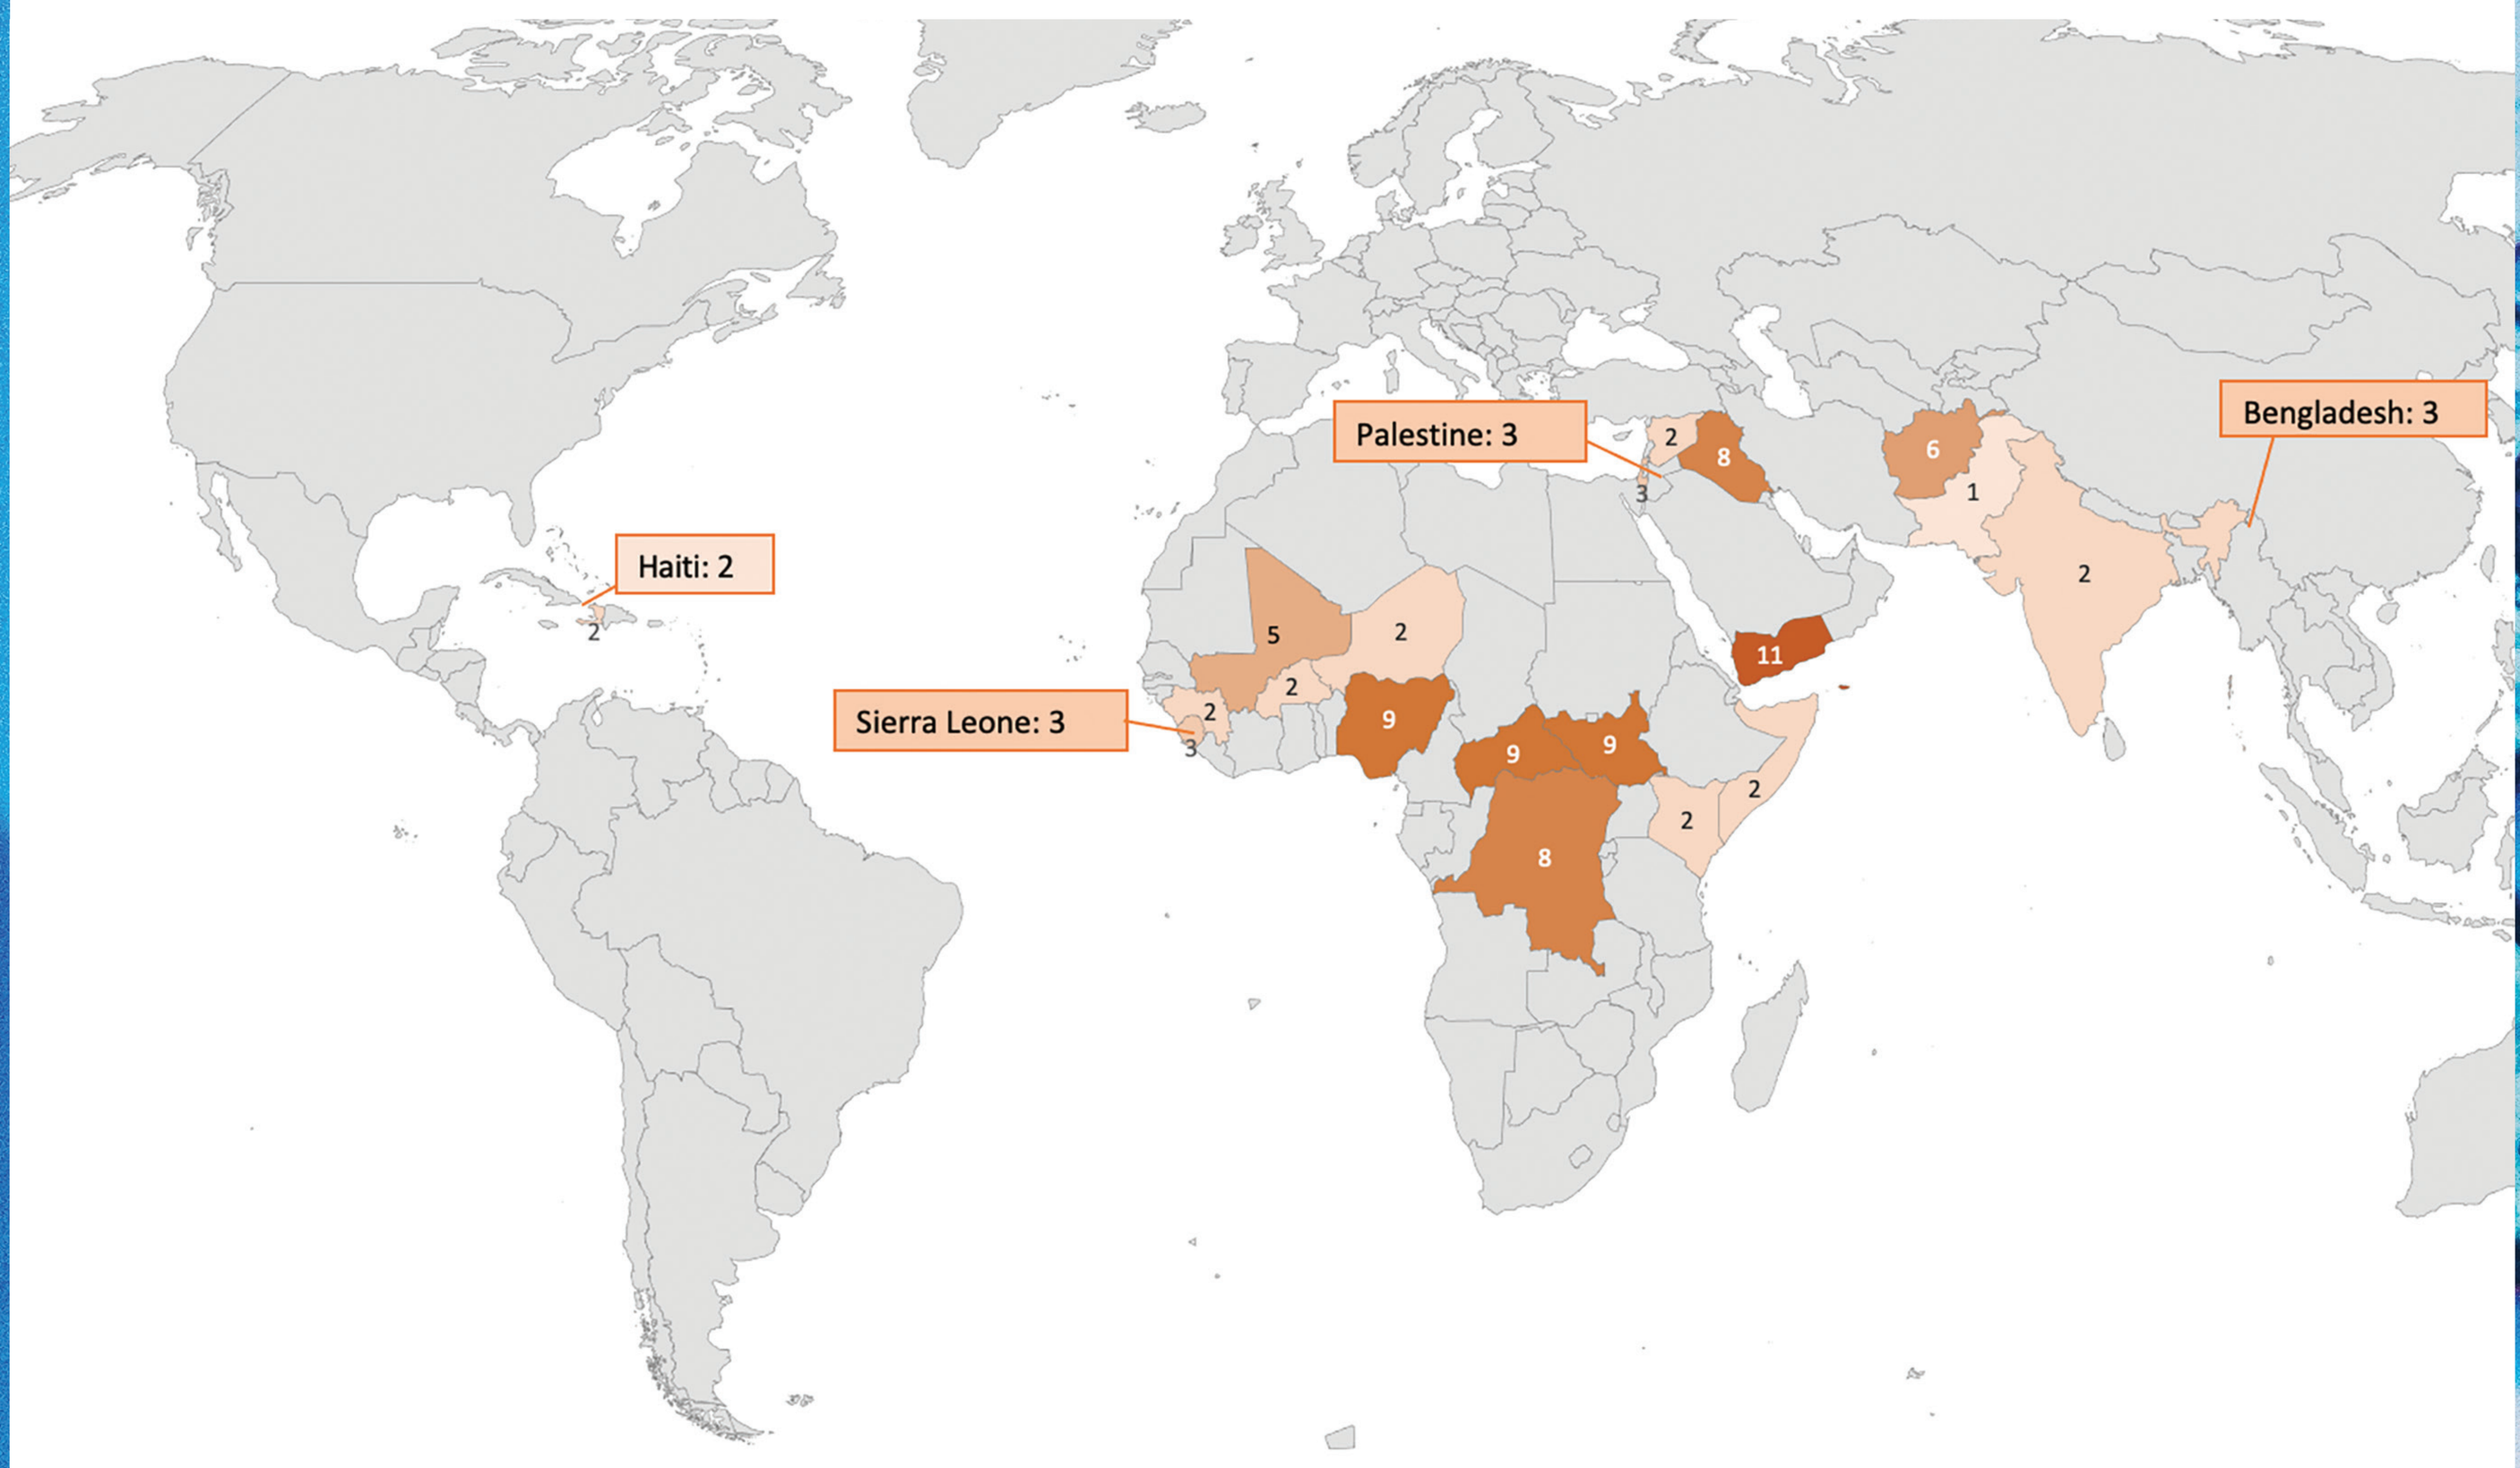
Palestine: 3
Bengladesh: 3
Haiti: 2
Sierra Leone: 3

These 9 months courses include 5 modules, work-based assignments and incorporates individual and group clinical mentorship provided by experienced professionals.

AIM

To assess the effectiveness of these courses in building competencies in AMS and IPC management, enhancing participants' confidence in performing IPC or AMS skills, and integrating it into their daily work practices.

METHODS

85 participants out of the 91 enrolled in the first & second cohorts completed the course between July 2021 and February 2023.

- Knowledge but also technical know-how (through skills confidence self-assessment) evaluated during pre- and post-course **Competency Gap Assessments**.
- **Participant feedback** on course content and pedagogical methods collected along the course.
- Implemented AMS or IPC interventions, obstacles faced and recommendations were gathered through electronic **surveys sent 6 months post-graduation** to participants, their line managers and headquarters technical advisors.

RESULTS

- Self-reported **CONFIDENCE LEVELS** to perform **SKILLS** in implementing AMS and IPC interventions substantially increased, with averages rising from 41% to **94%**, and 57% to **96%** respectively.
- The 6 months post-course survey revealed successful **IMPLEMENTATION** of AMS and IPC **ACTIVITIES**:

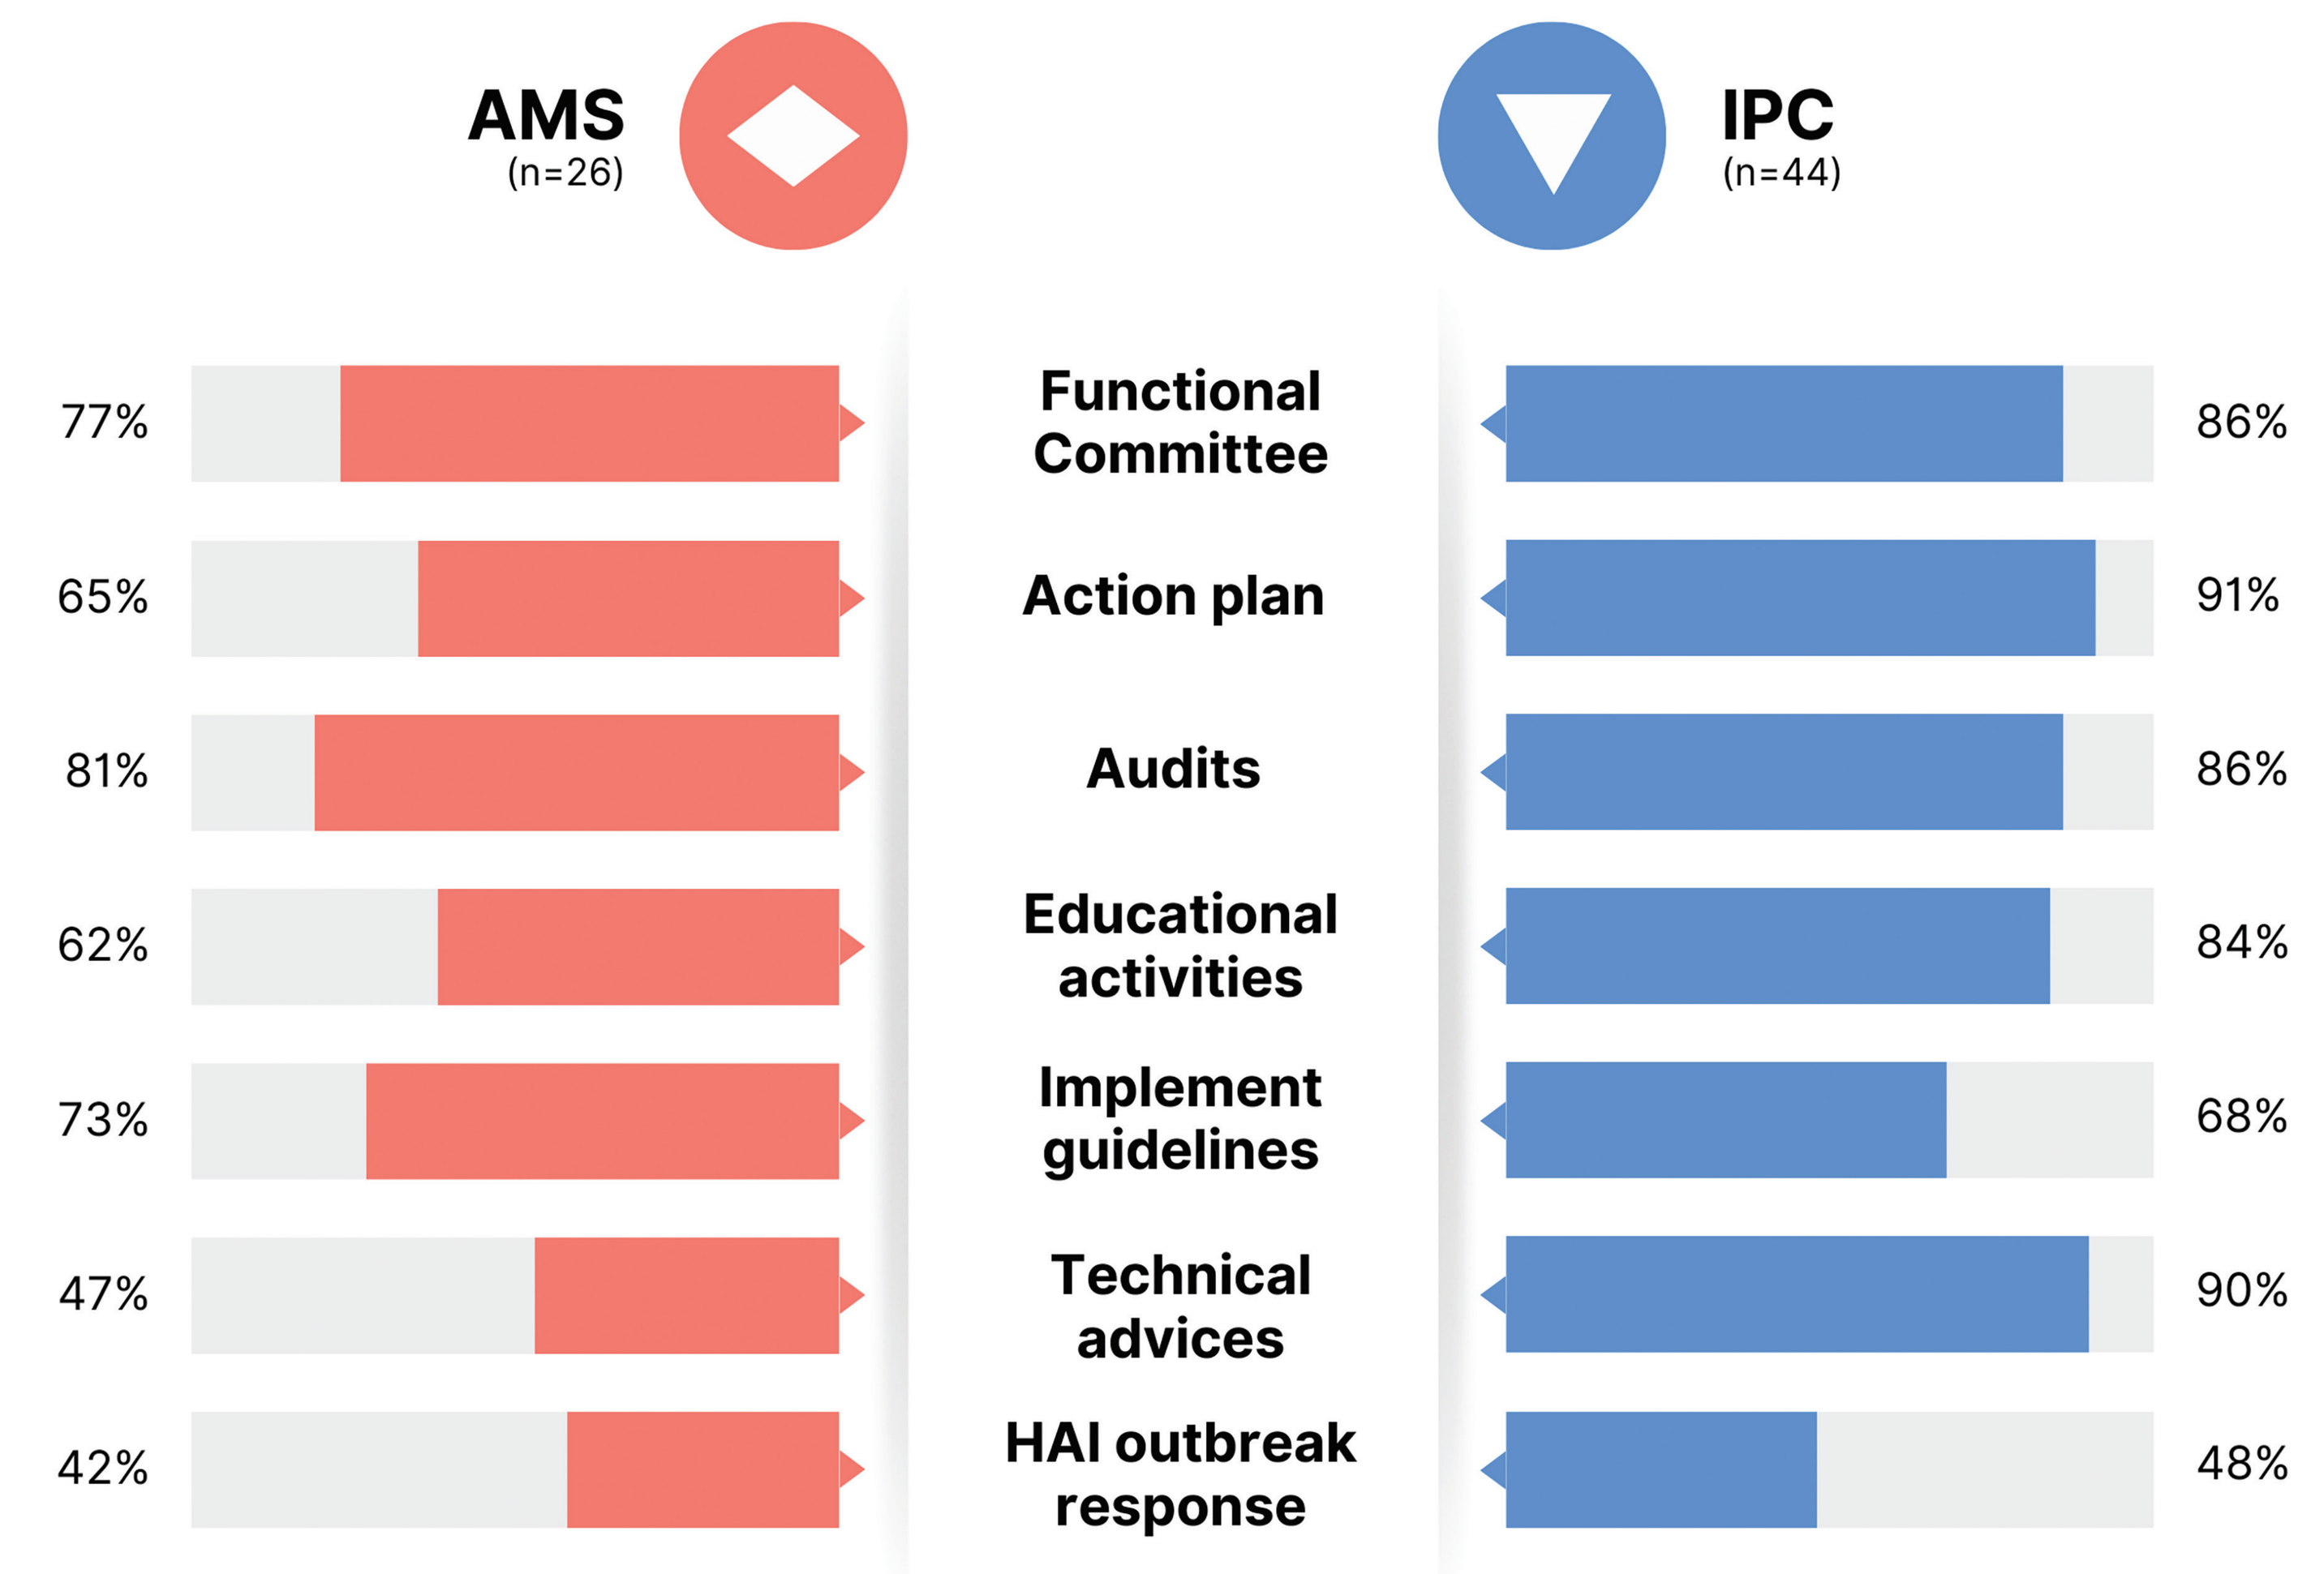
AMS
(n=26)
IPC
(n=44)
77% Functional Committee 86%
65% Action plan 91%
81% Audits 86%
62% Educational activities 84%
73% Implement guidelines 68%
47% Technical advices 90%
42% HAI outbreak response 48%

- Insufficient drugs and limited human resources were the **MAIN OBSTACLES** in AMS implementation, while IPC faced challenges primarily due to resistance to behavior change and inadequate infrastructure.
- The most **IMPACTFUL LEARNING ACTIVITIES** fed-back by the participants was the mentoring, either in individual (100%) or in group session (99%):

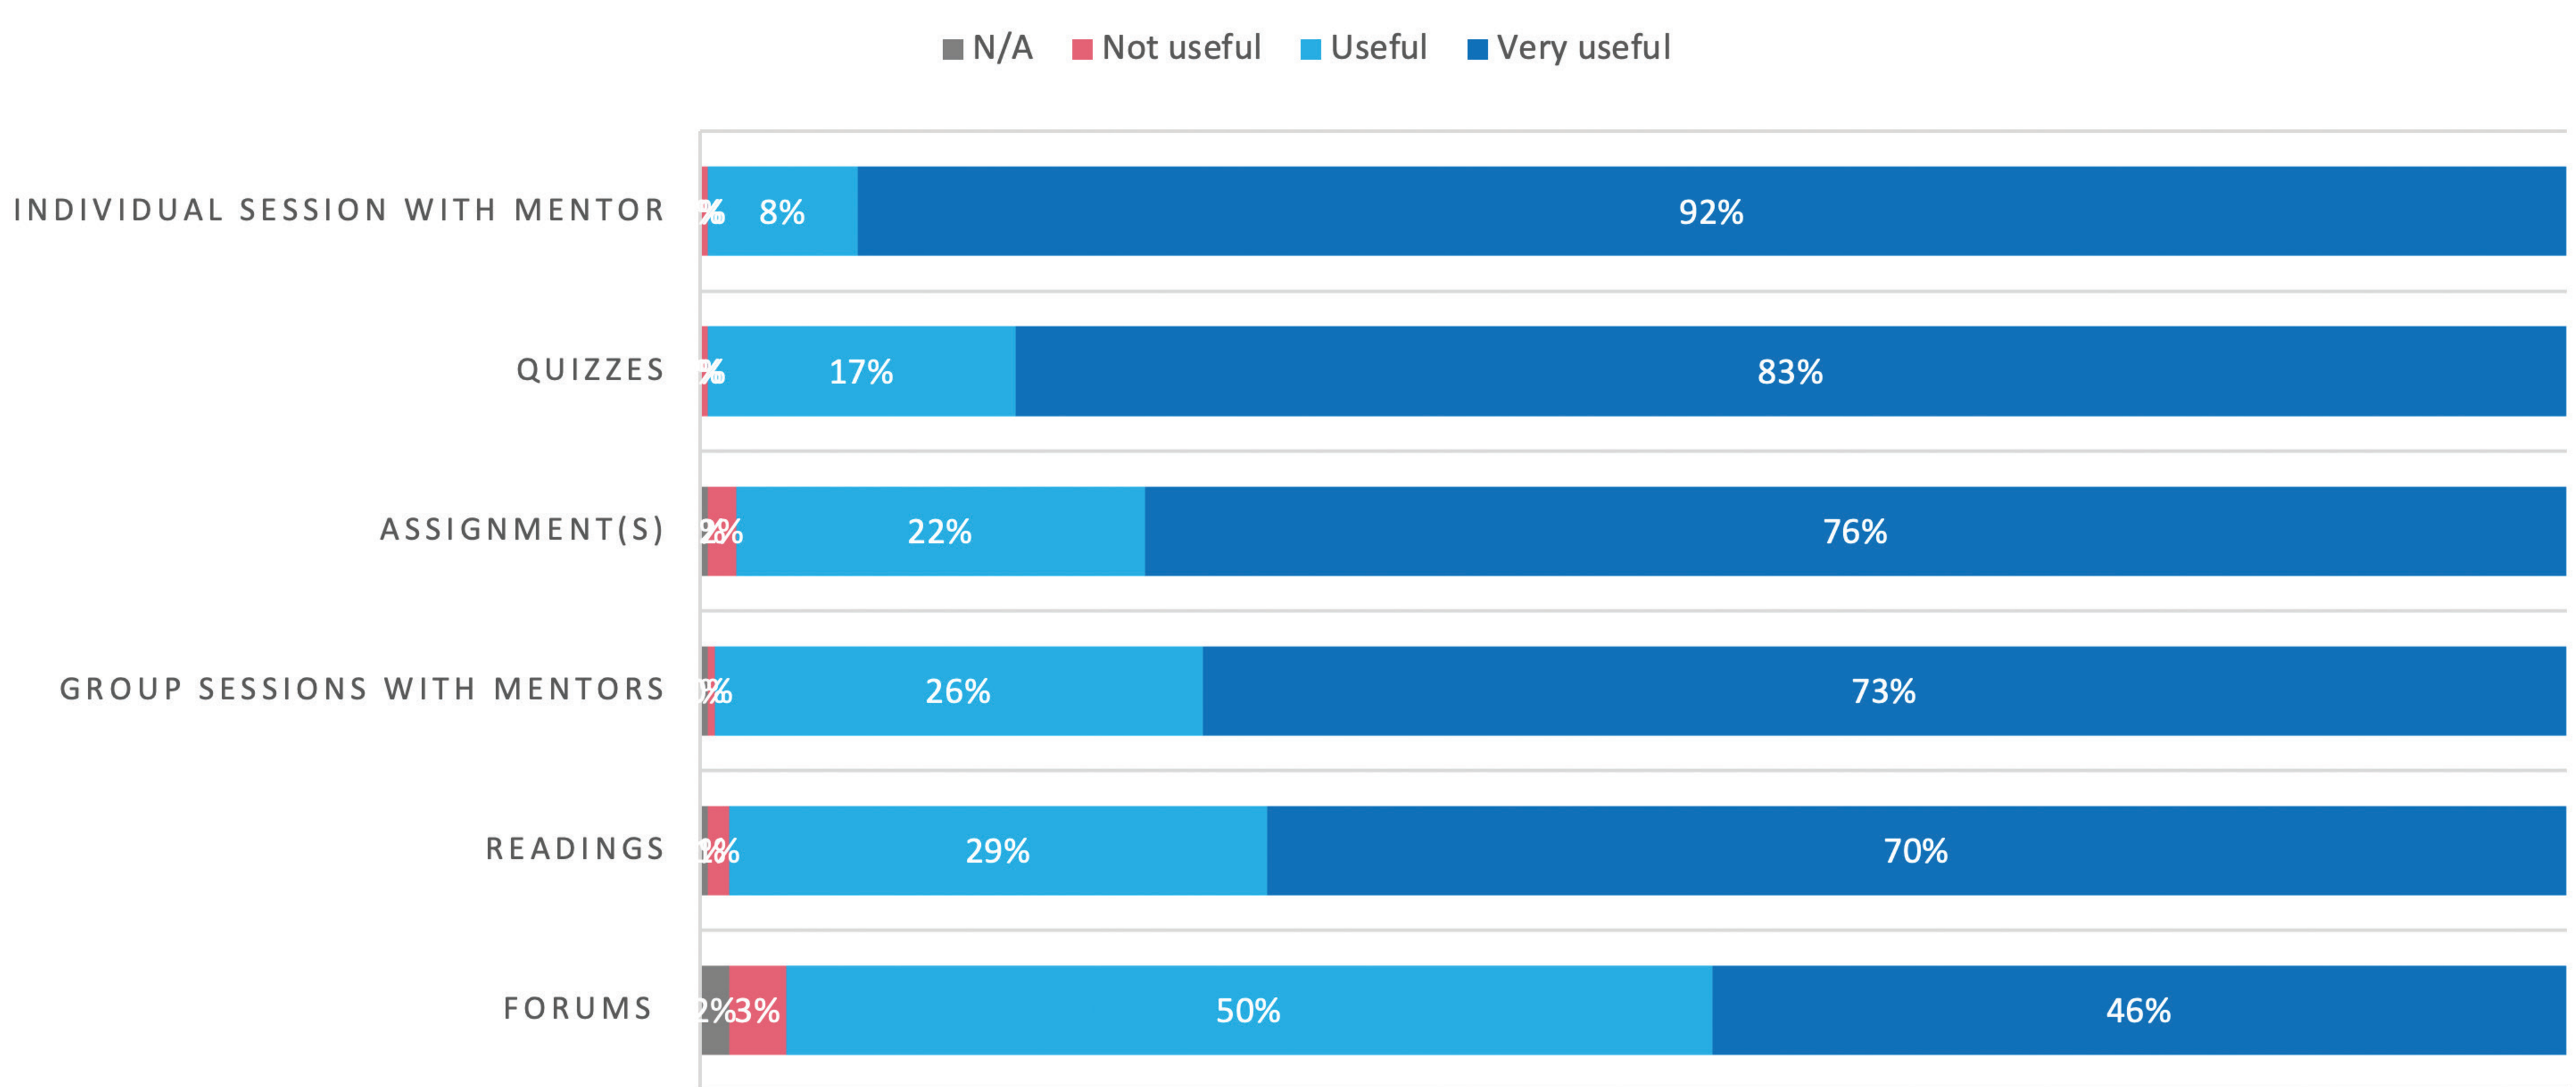
N/A
Not useful
Useful
Very useful
INDIVIDUAL SESSION WITH MENTOR
8%
92%
QUIZZES
17%
83%
ASSIGNMENT(S)
22%
76%
GROUP SESSIONS WITH MENTORS
26%
73%
READINGS
29%
70%
FORUMS
2%
3%
50%
46%

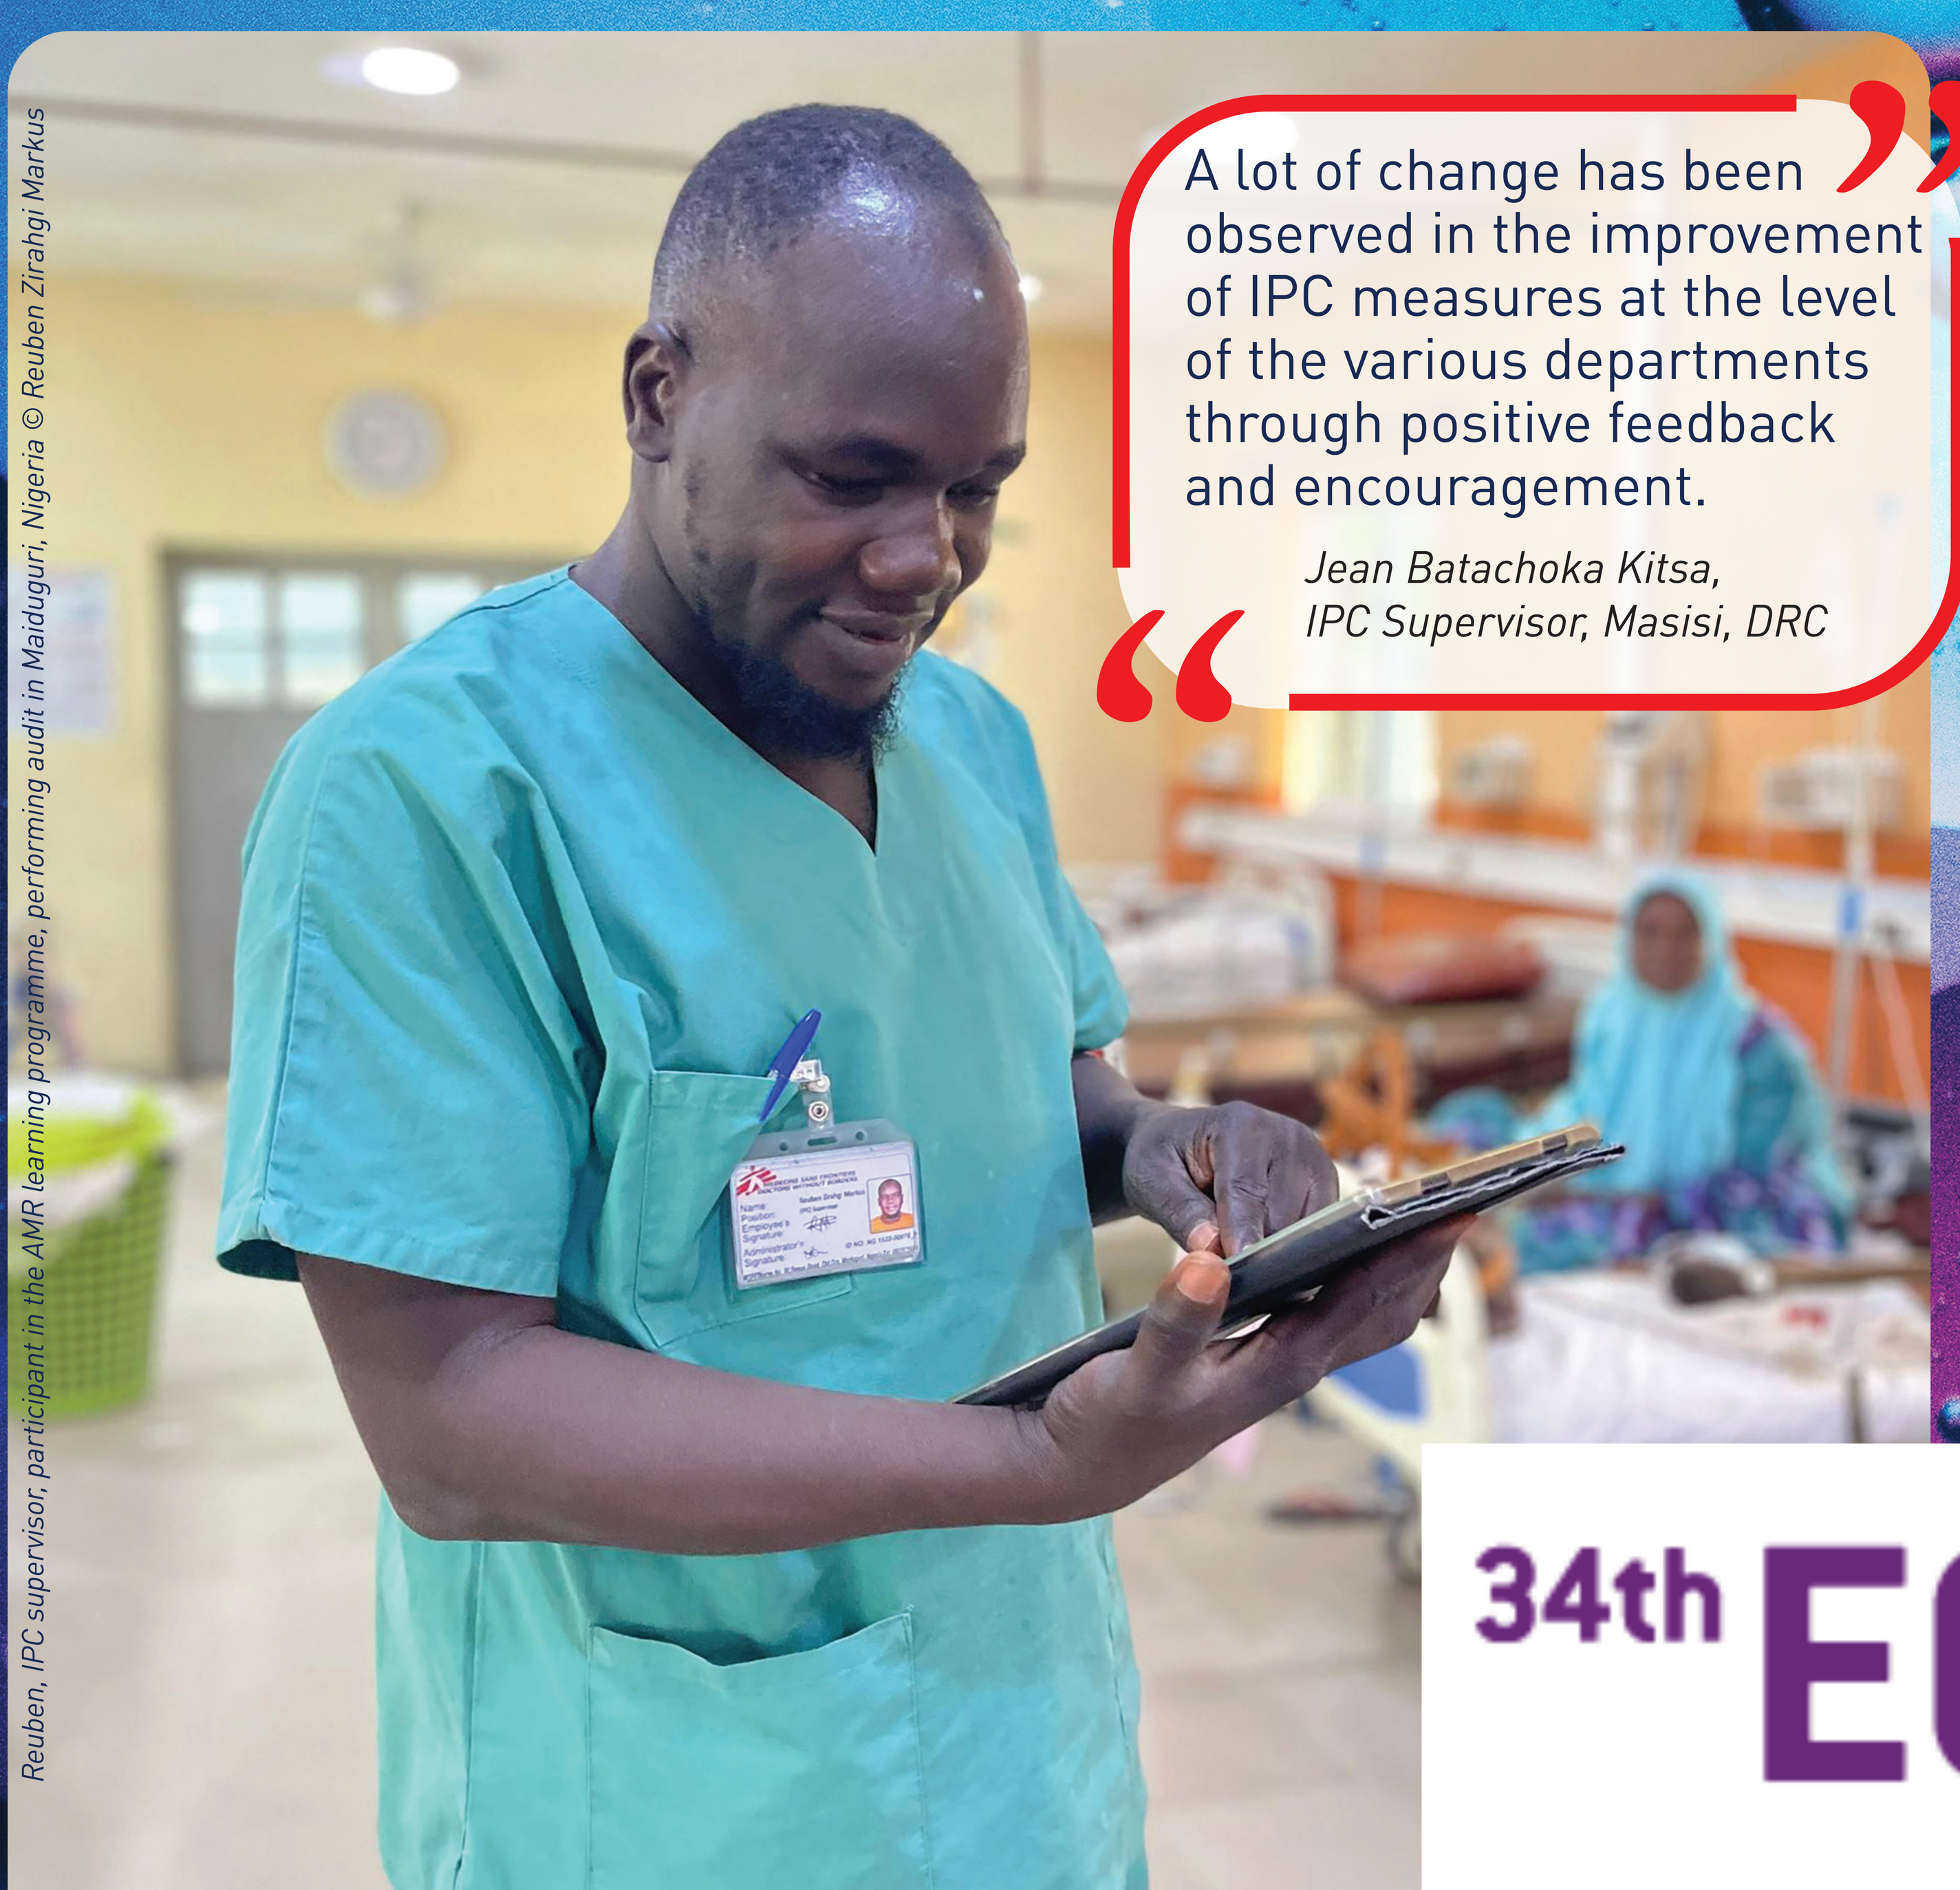
A lot of change has been observed in the improvement of IPC measures at the level of the various departments through positive feedback and encouragement.
Jean Batachoka Kitsa,
IPC Supervisor, Masisi, DRC
Reuben, IPC supervisor, participant in the AMR learning programme, performing audit in Maiduguri, Nigeria © Reuben Zirahgi Markus

CONCLUSION

By **combining e-learning with clinical mentorship**, the courses effectively bridge theoretical knowledge and practical application. Despite the reported obstacles to implementation, this approach demonstrates a clear benefit: **increased skills confidence** among participants and a **boost in AMS & IPC interventions** in the working environment.

The findings highlight the pivotal role of the AMR learning initiative in supporting and strengthening AMS and IPC capacity and expertise on a global scale, therefore contributing to enhance healthcare quality and reduce the risk of AMR transmission.

ACKNOWLEDGEMENT

Our deepest gratitude to MSF advisors, British Society for Antimicrobial Chemotherapy for their invaluable support and collaboration. We also thank our mentors and participants for enriching contributions and commitment to improving IPC/AMS in healthcare.

34th ECCMID EUROPEAN CONGRESS OF CLINICAL MICROBIOLOGY AND INFECTIOUS DISEASES

Barcelona, Spain
27 – 30 April 2024